AAAAI Membership Experience with Allergen Immunotherapy Safety in Young Children and Pregnant Women

Matthew A. Rank, MD[1]; David W. Hauswirth, MD[2]; Christopher W. Calabria, MD[3]; Lawrence D. Sher, MD[4]; Desiree Larenas Linnemann, MD[5]

[1]Mayo Clinic – Scottsdale, AZ; [2]The Ohio State University – Columbus, OH; [3]Dilley Allergy and Asthma – Helotes, TX; [4]Peninsula Research Associates – Rolling Hills Estates, CA; [5]Hospital Medica Sur – Mexico City, Mexico

Background

Rationale: There are limited safety data for allergen immunotherapy (AIT) in children and pregnant women; therefore, surveying current practice patterns may provide important preliminary guidance.

Methods: A web-based survey was sent out to AAAAI members in August 2012 and consisted of 9 questions, with an optional 32 additional questions. Major problems with AIT were defined as activation of an underlying disease, systemic reactions, or discontinuation of AIT for medical reasons. Minor problems with AIT were defined as dose-reduction or postponed dose and AIT continuation. Survey results are expressed using descriptive statistics.

Results: Of 5,148 requests to complete the survey, 1,061 (20.6%) were returned. Survey responder demographics were: 86% US-based, 31% in an academic setting, and 54% with >16 years of experience. Usual starting age for AIT was 5% at age 2, 9% age 3, 14% age 4, and 48% for age 5. Eighty-three percent had continued AIT in pregnant women while only 3% felt that to be contraindicated. Based on a total experience of > 3829 pregnant women who continued AIT during pregnancy, 2% report major problems and 22% minor problems. Ninety-five percent had not started AIT in pregnant women. Based on a total experience of >164 pregnant women who started AIT during pregnancy, 6% report major problems and 9% report minor problems.

Conclusions: A survey of AAAAI members suggests that most providers begin AIT in children by age 5, most continue AIT in pregnant women and rarely report major problems in this population, and very few members start AIT in pregnant women.

Rationale

- Limited safety data for AIT in very young children and pregnant women
- Surveying current practice patterns may provide guidance

Methods

- Web-based survey was sent out to AAAAI members in August 2012 and consisted of 9 questions, with an optional 32 additional questions
- Survey results are expressed using descriptive statistics

Definitions

- **Major problems** with AIT were defined as activation of an underlying disease, systemic reactions, or discontinuation of AIT for medical reasons
- **Minor problems** with AIT were defined as dose-reduction or postponed dose and AIT continuation

Results

- Response rate: 20% (1,016 of 5,148)

Figure 1: Responder Practice Location

Where do you practice?

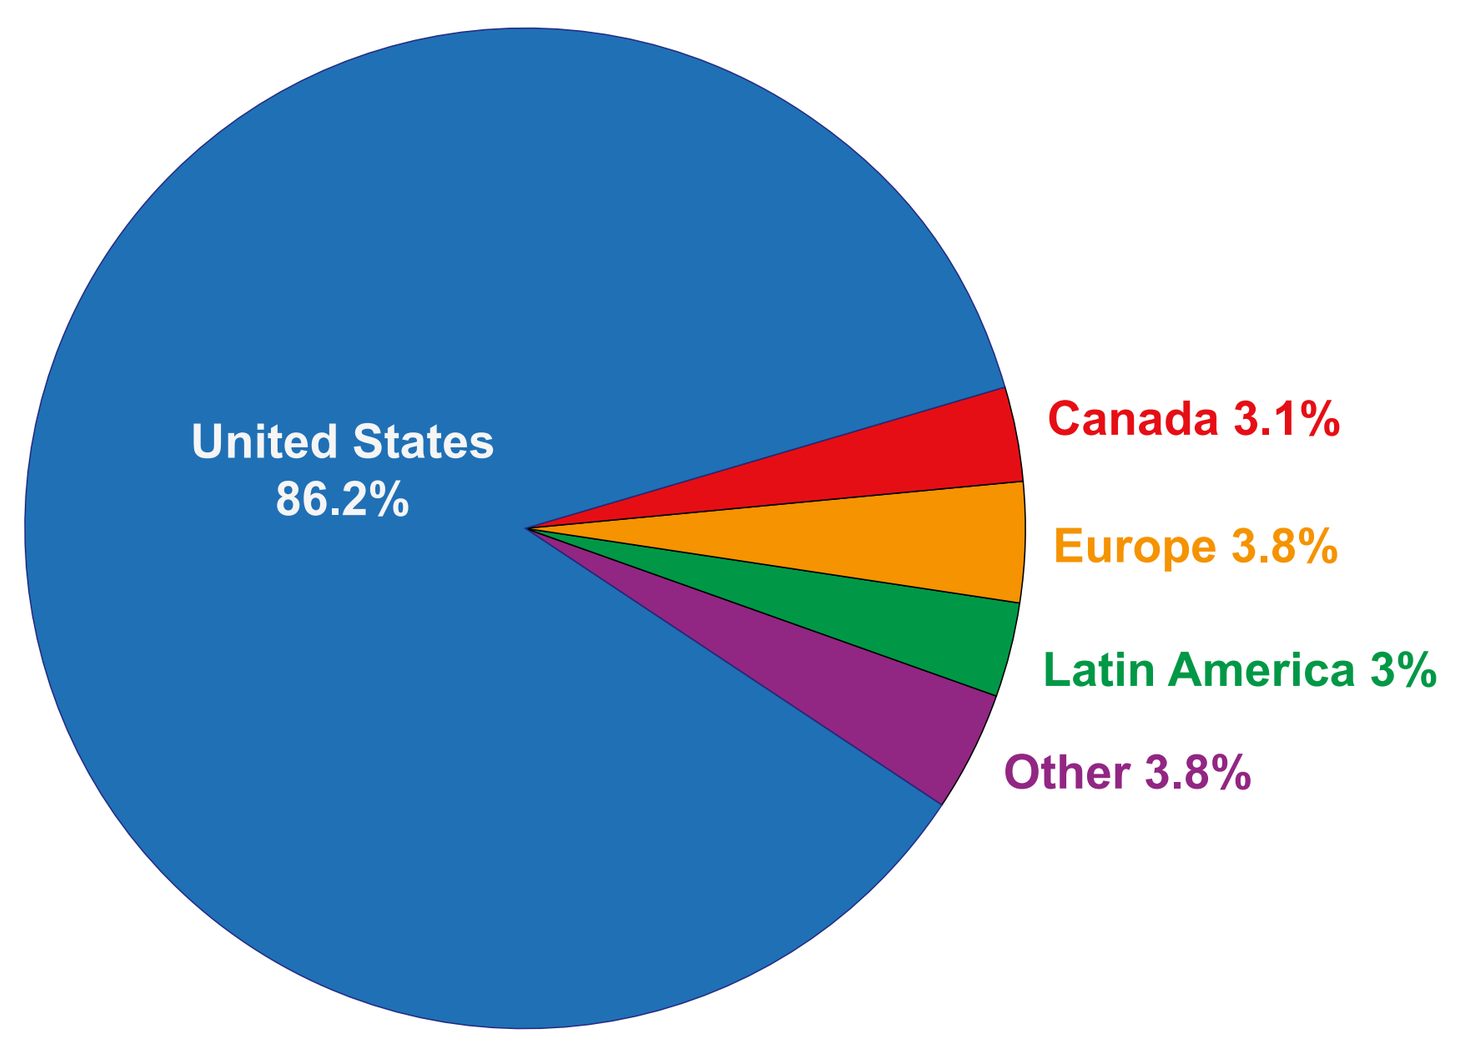
United States
86.2%
Canada 3.1%
Europe 3.8%
Latin America 3%
Other 3.8%

Figure 2: Responder Practice Setting

In what setting do you practice?

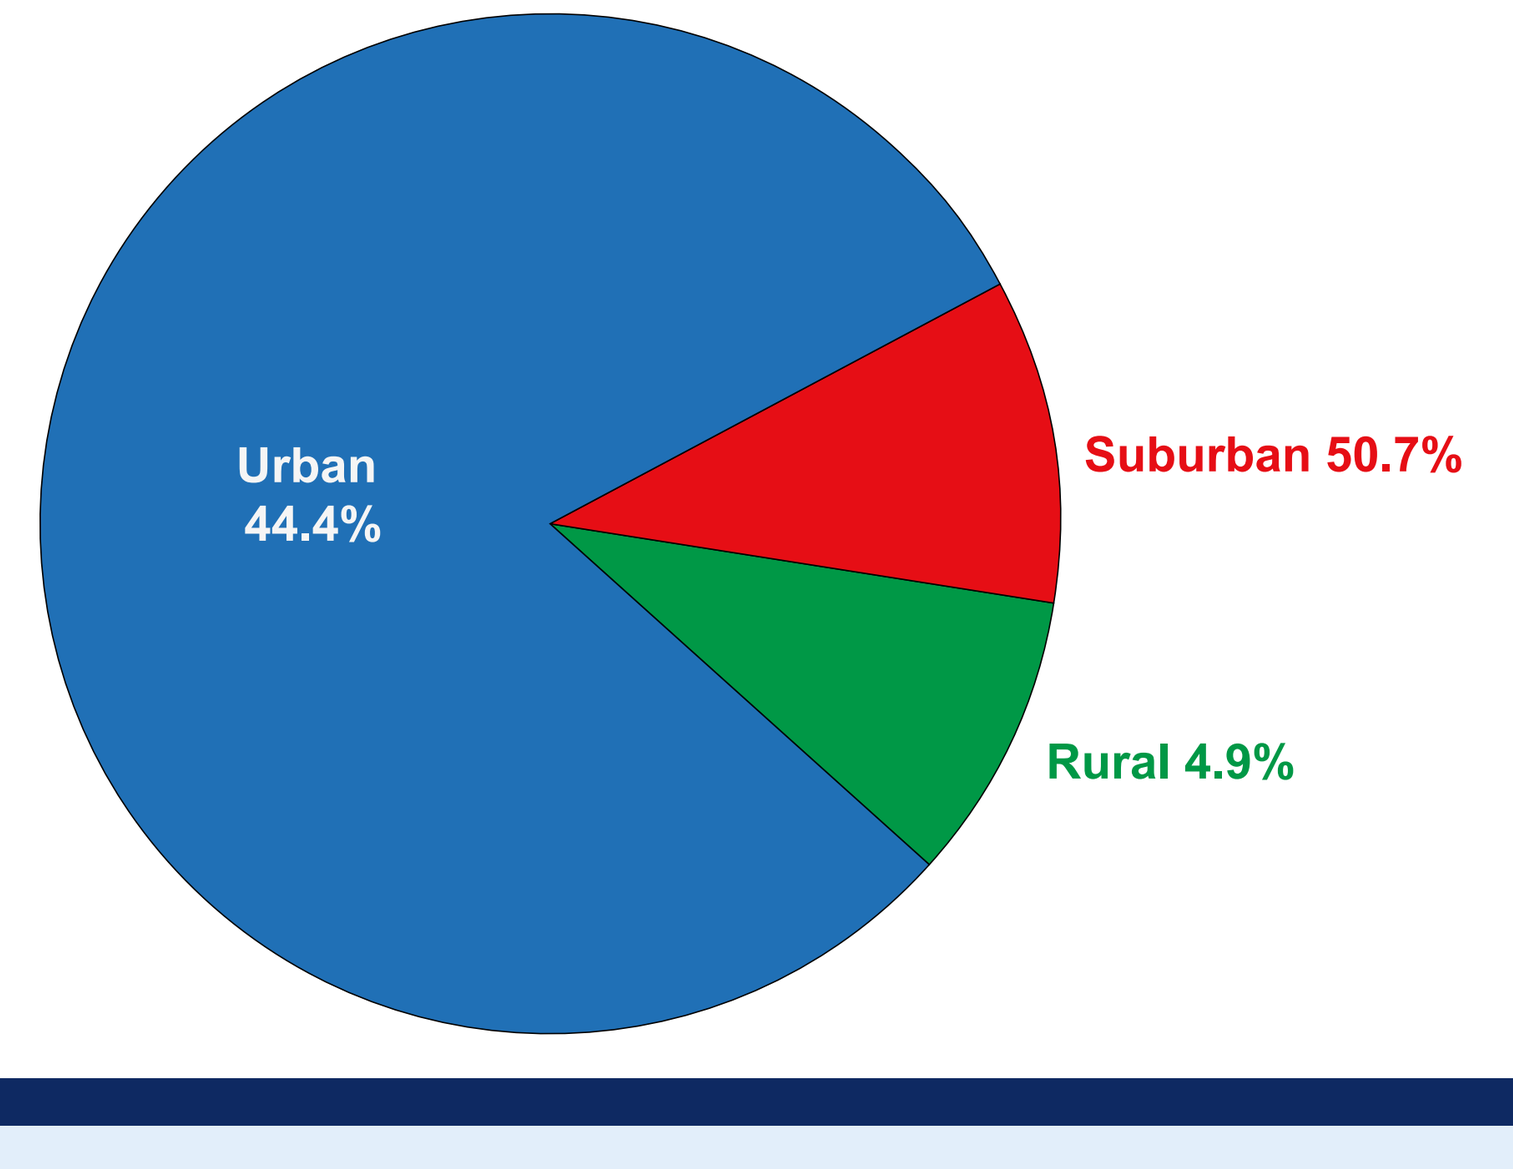
Urban
44.4%
Suburban 50.7%
Rural 4.9%

Figure 3: Responder Practice Type

Do you practice in an academic setting?

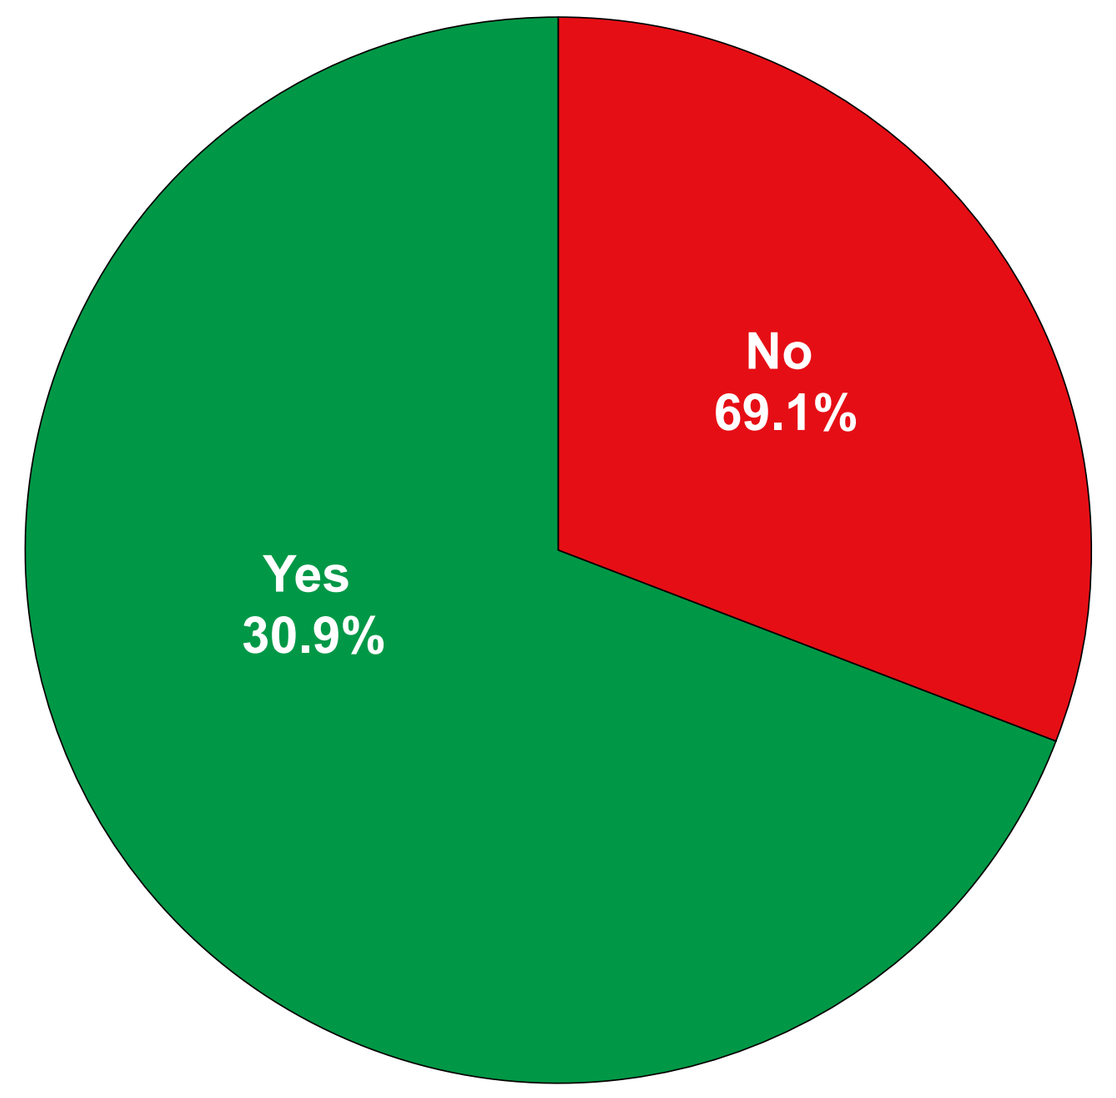
No
69.1%
Yes
30.9%

Figure 4: Responder Years of Experience

How many years have you been in practice an as allergist?

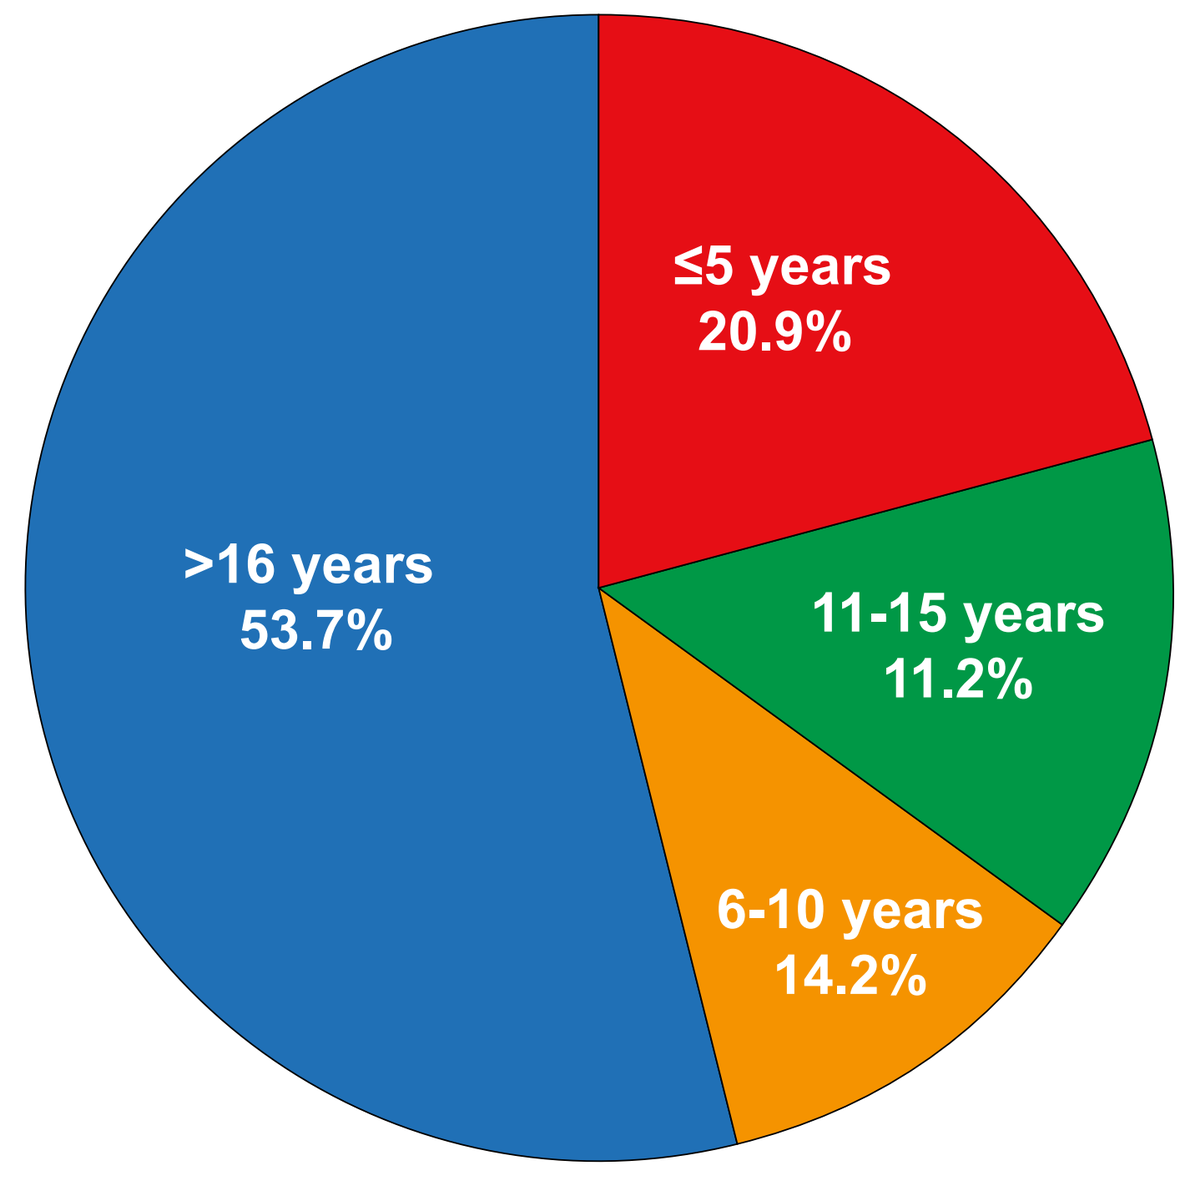
≤5 years
20.9%
>16 years
53.7%
11-15 years
11.2%
6-10 years
14.2%

Figure 5: Responder Experience with AIT

How many patients do you have on immunotherapy or are treated with immunotherapy at this moment in your clinic (shots can be give in your clinic or elsewhere but under your management)?

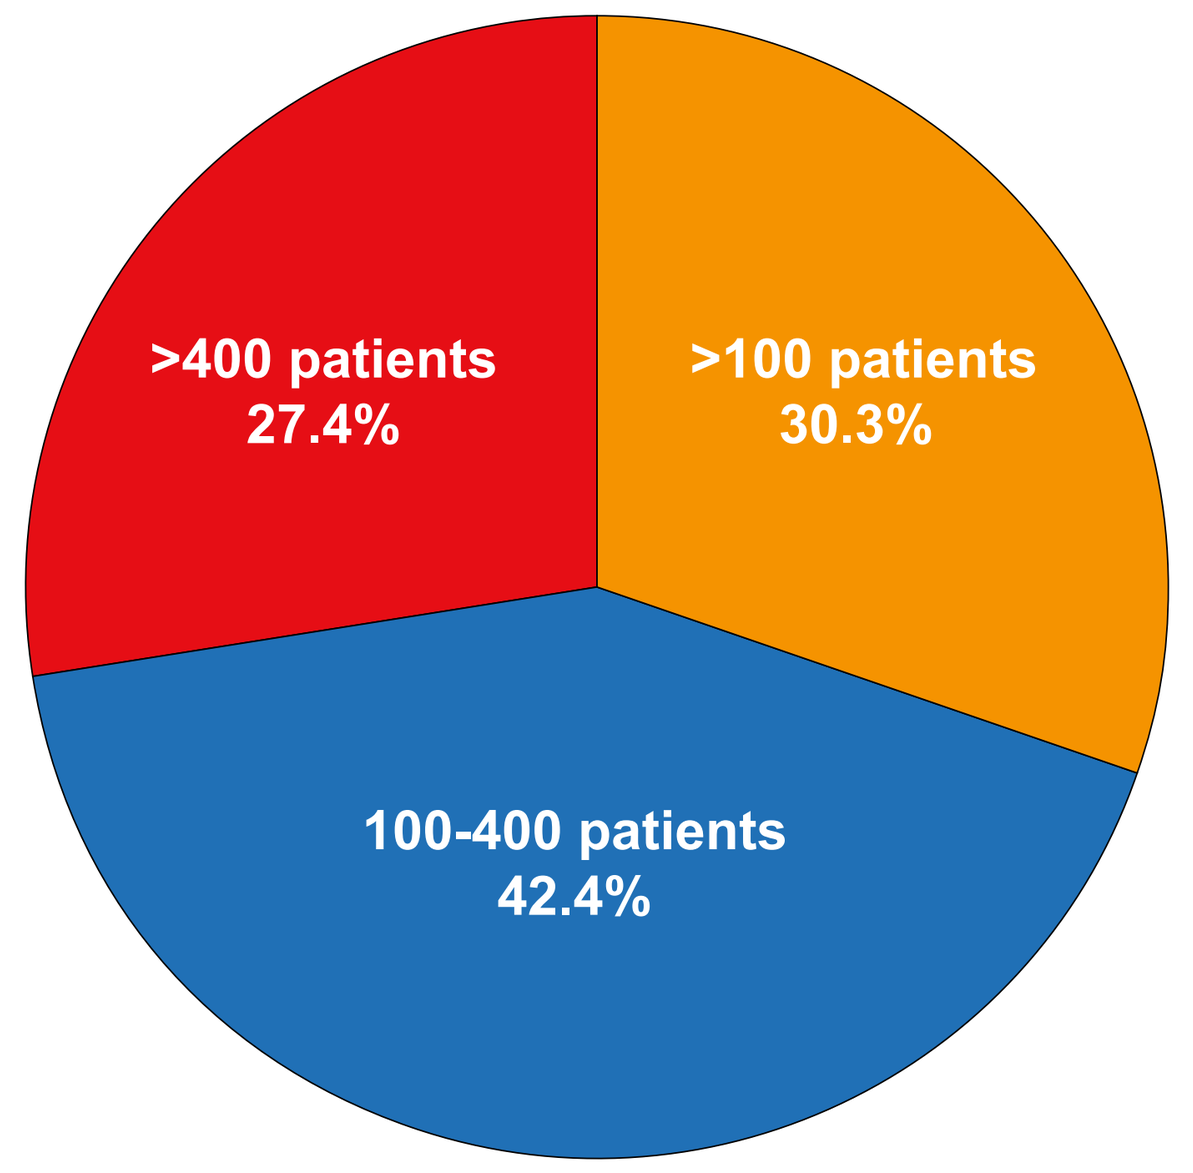
>400 patients
27.4%
>100 patients
30.3%
100-400 patients
42.4%

Figure 6: Responder AIT Dosing

In general, do you consider yourself as managing what dose:

Low: below practice parameters recommended doses

Medium: at lower range of practice parameters recommended doses

High: at higher range of practice parameters recommended doses or above

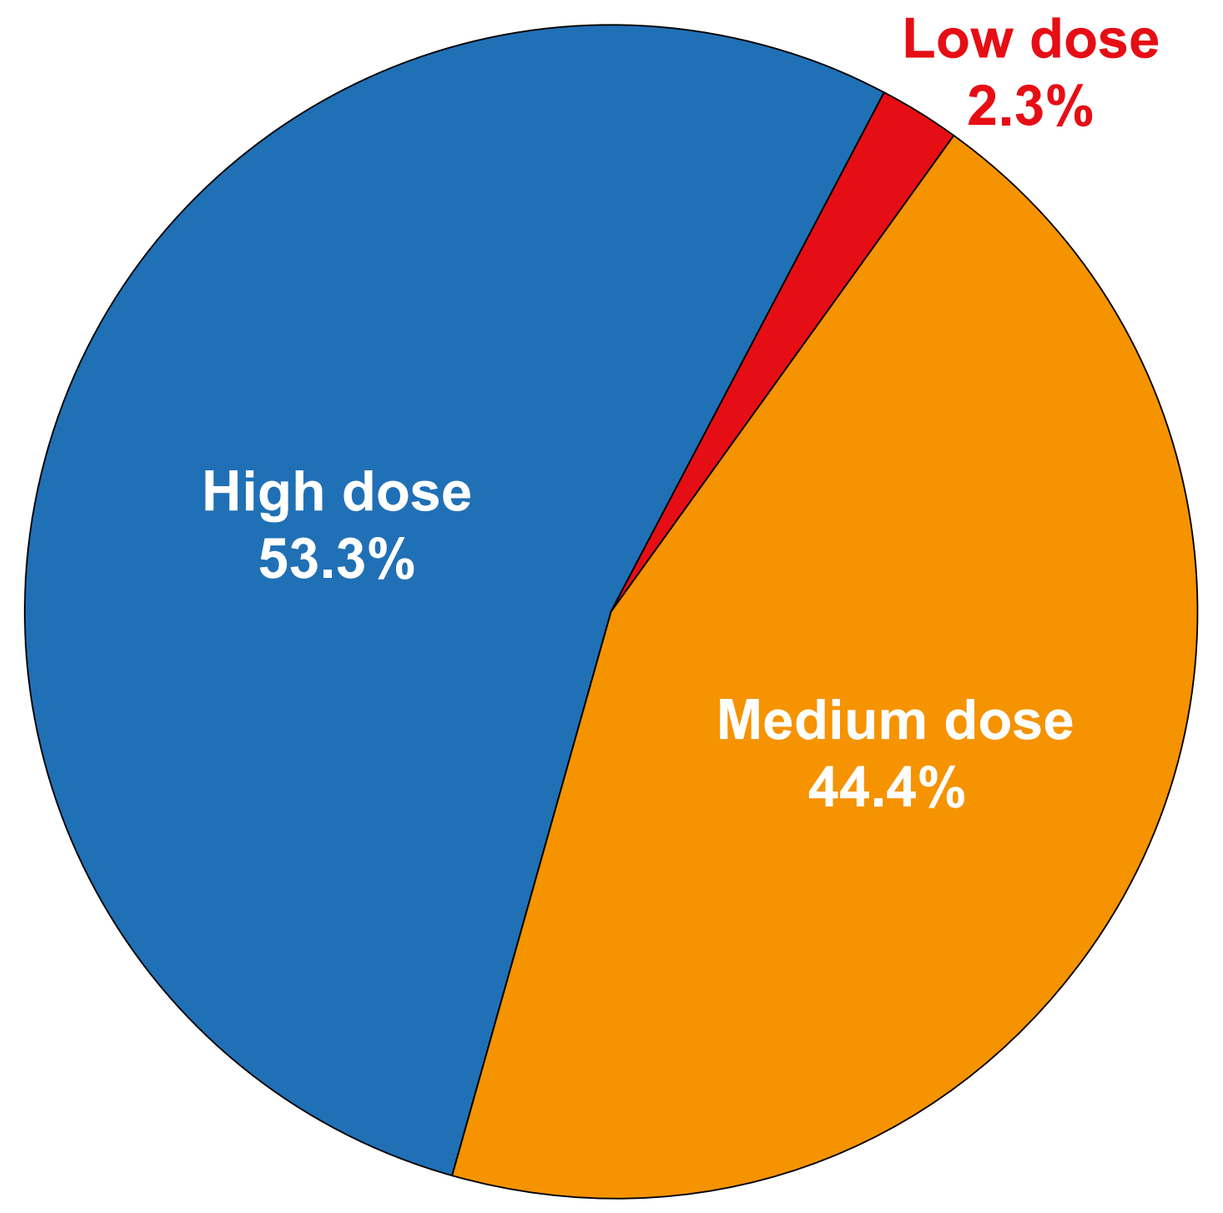
Low dose
2.3%
High dose
53.3%
Medium dose
44.4%

Figure 7: Starting age of AIT

From what age onward do you give allergen immunotherapy (not venom) to children?

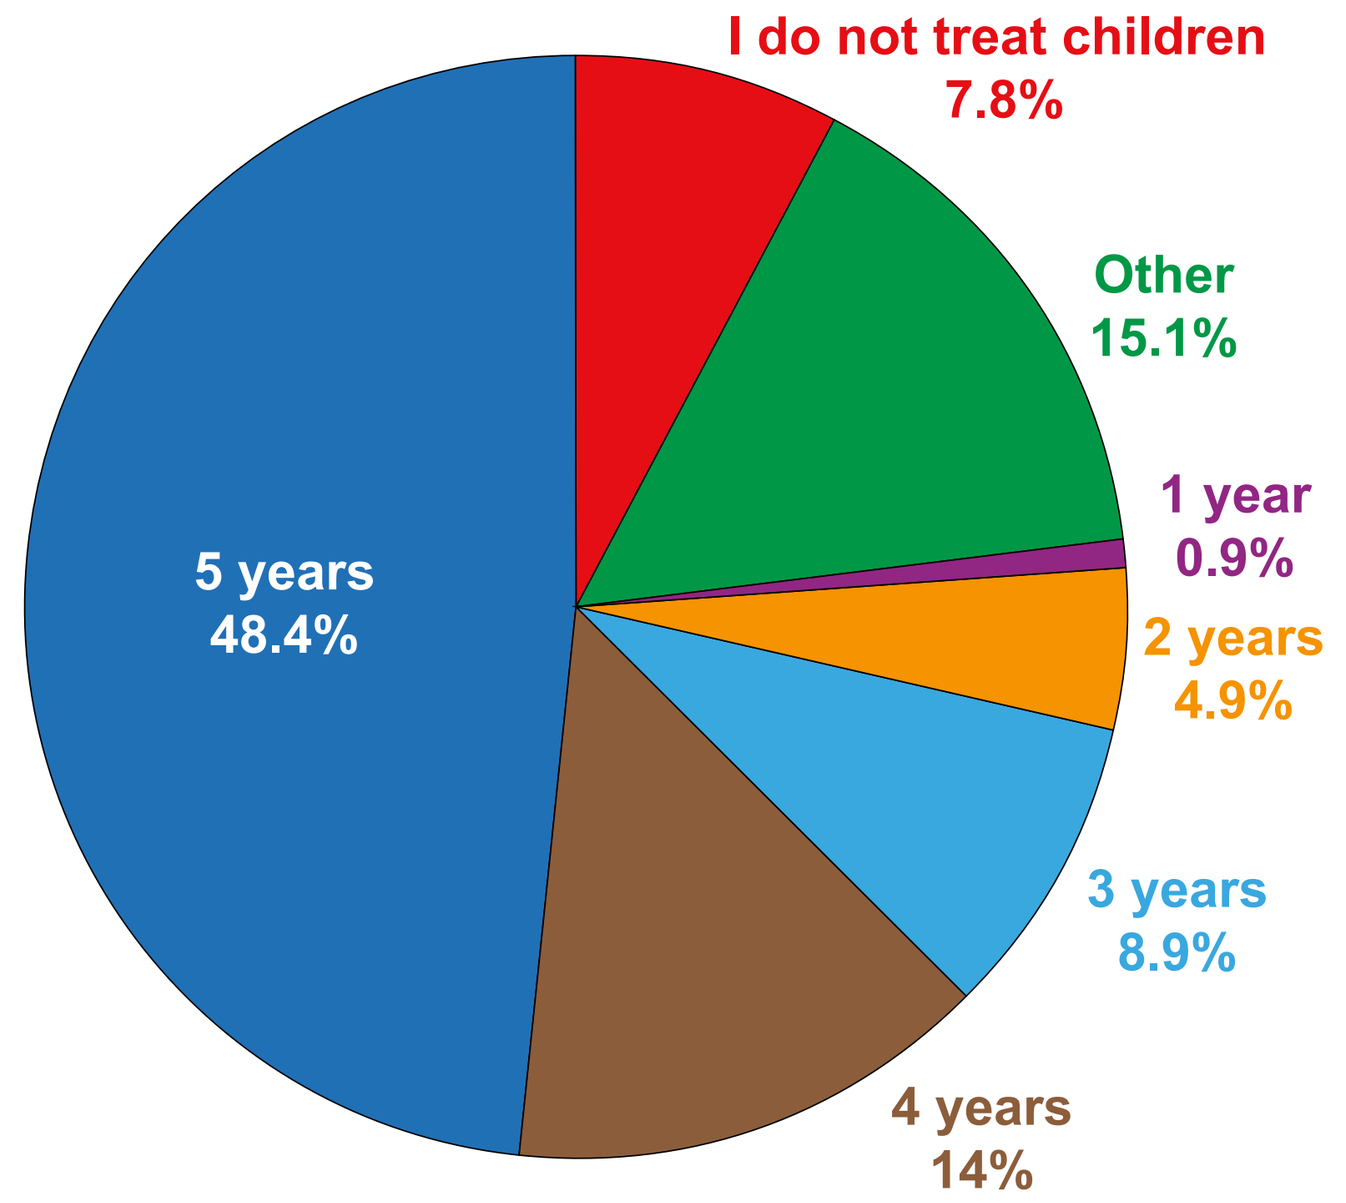
I do not treat children
7.8%
Other
15.1%
1 year
0.9%
2 years
4.9%
3 years
8.9%
4 years
14%
5 years
48.4%

Figure 8: AIT in pregnancy

Have you had patients with the below medical conditions on AIT?

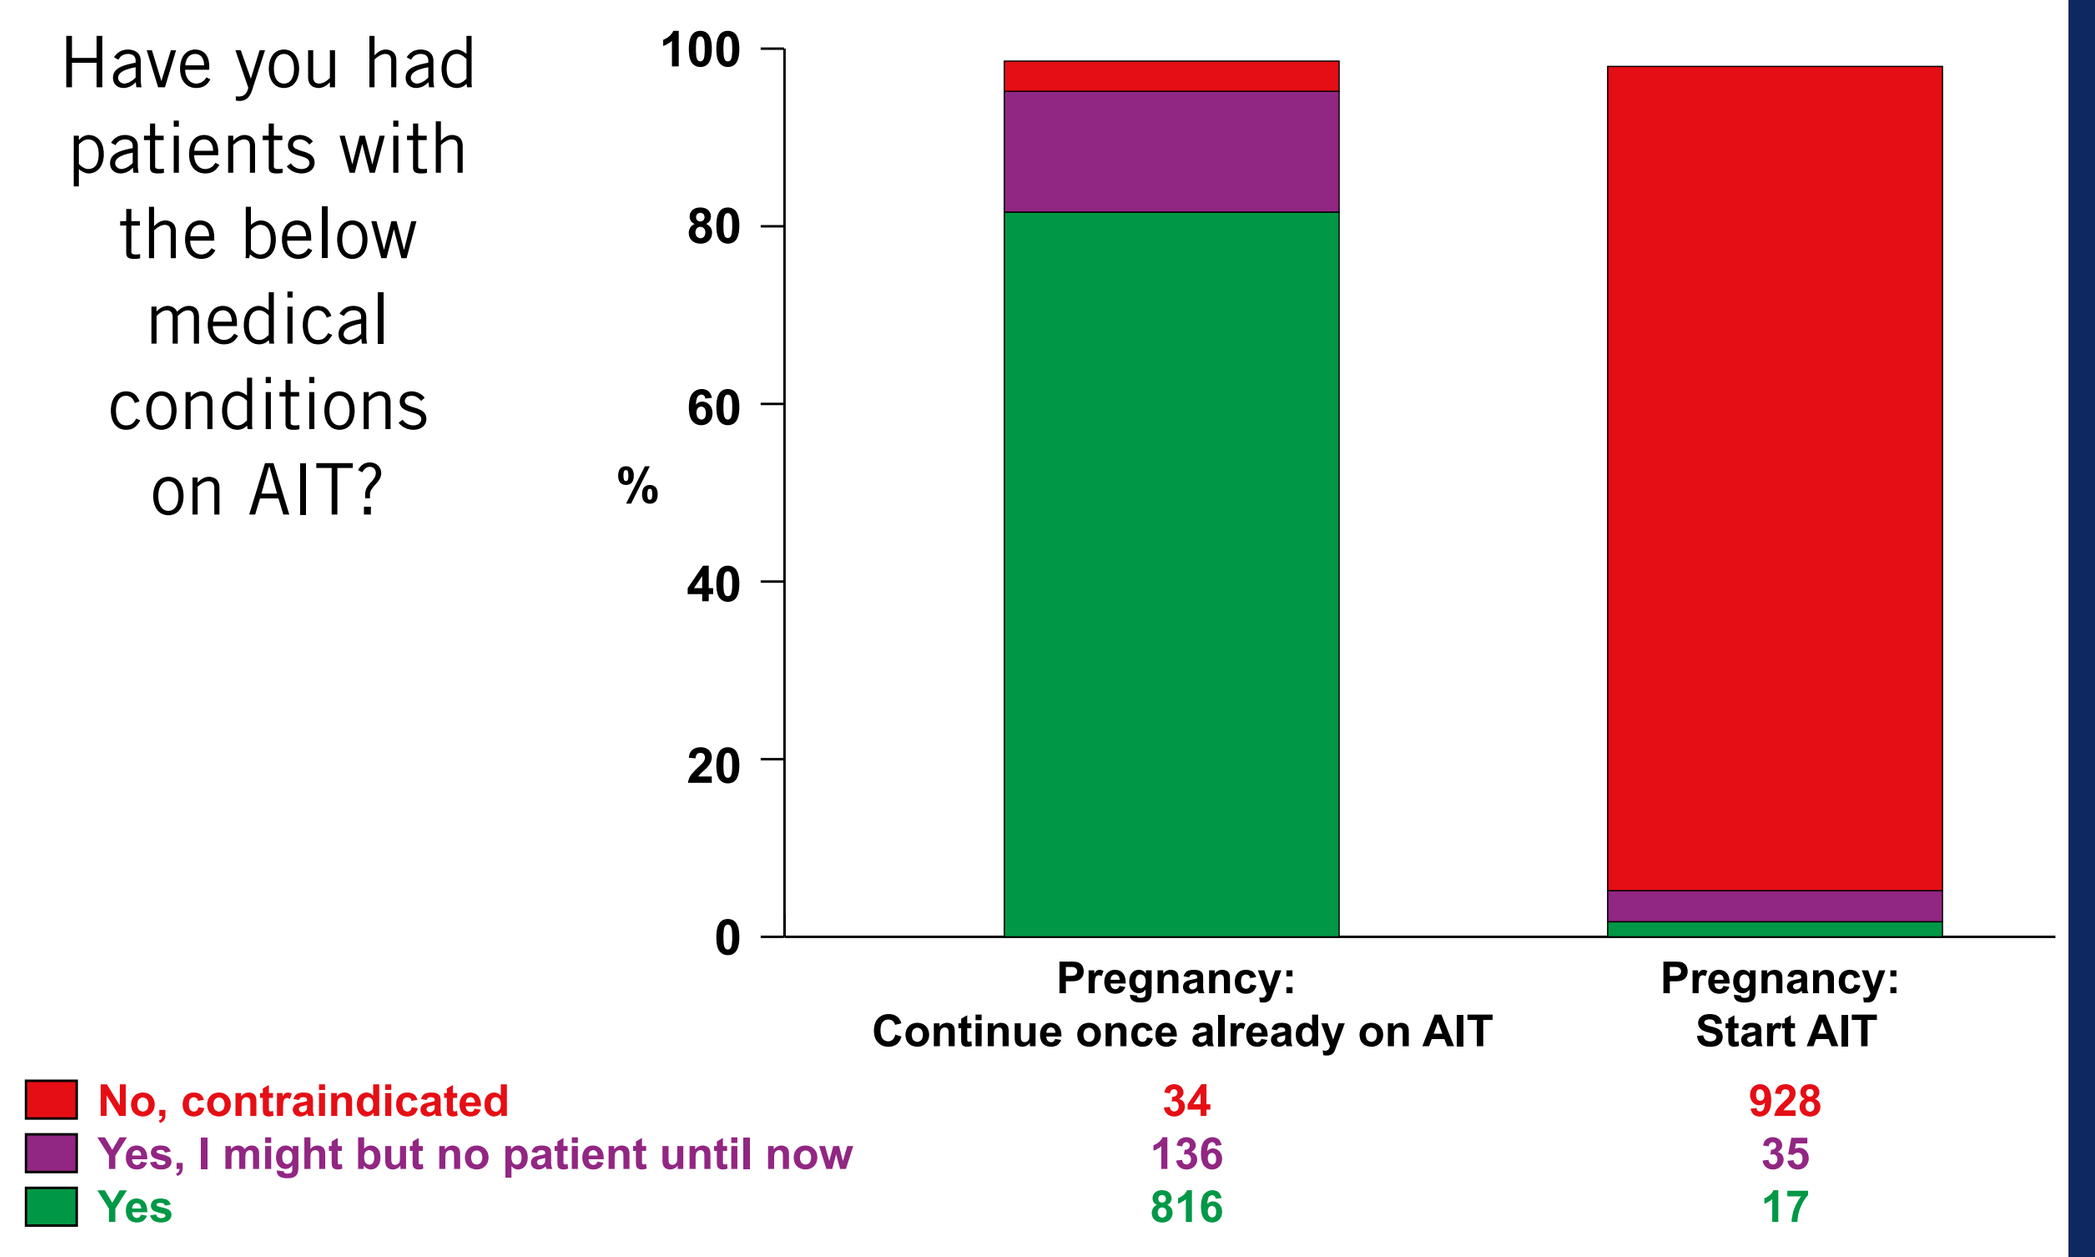
100
80
60
40
20
0
%
Pregnancy:
Continue once already on AIT
Pregnancy:
Start AIT
No, contraindicated 34 928
Yes, I might but no patient until now 136 35
Yes 816 17

Membership Experience: Pregnancy

- Continue AIT: Based on a total experience of >3,829 pregnant women who continued AIT during pregnancy:
 - 2% report major problems
 - 22% minor problems
- Start AIT: Based on a total experience of >164 pregnant women who started AIT during pregnancy:
 - 6% report major problems
 - 9% report minor problems

Conclusions

- Most providers begin AIT in children by age 5
- Most continue AIT in pregnant women and report a low rate of major problems in this population
- Very few members start AIT in pregnant women and those that have report 6% have a major problem

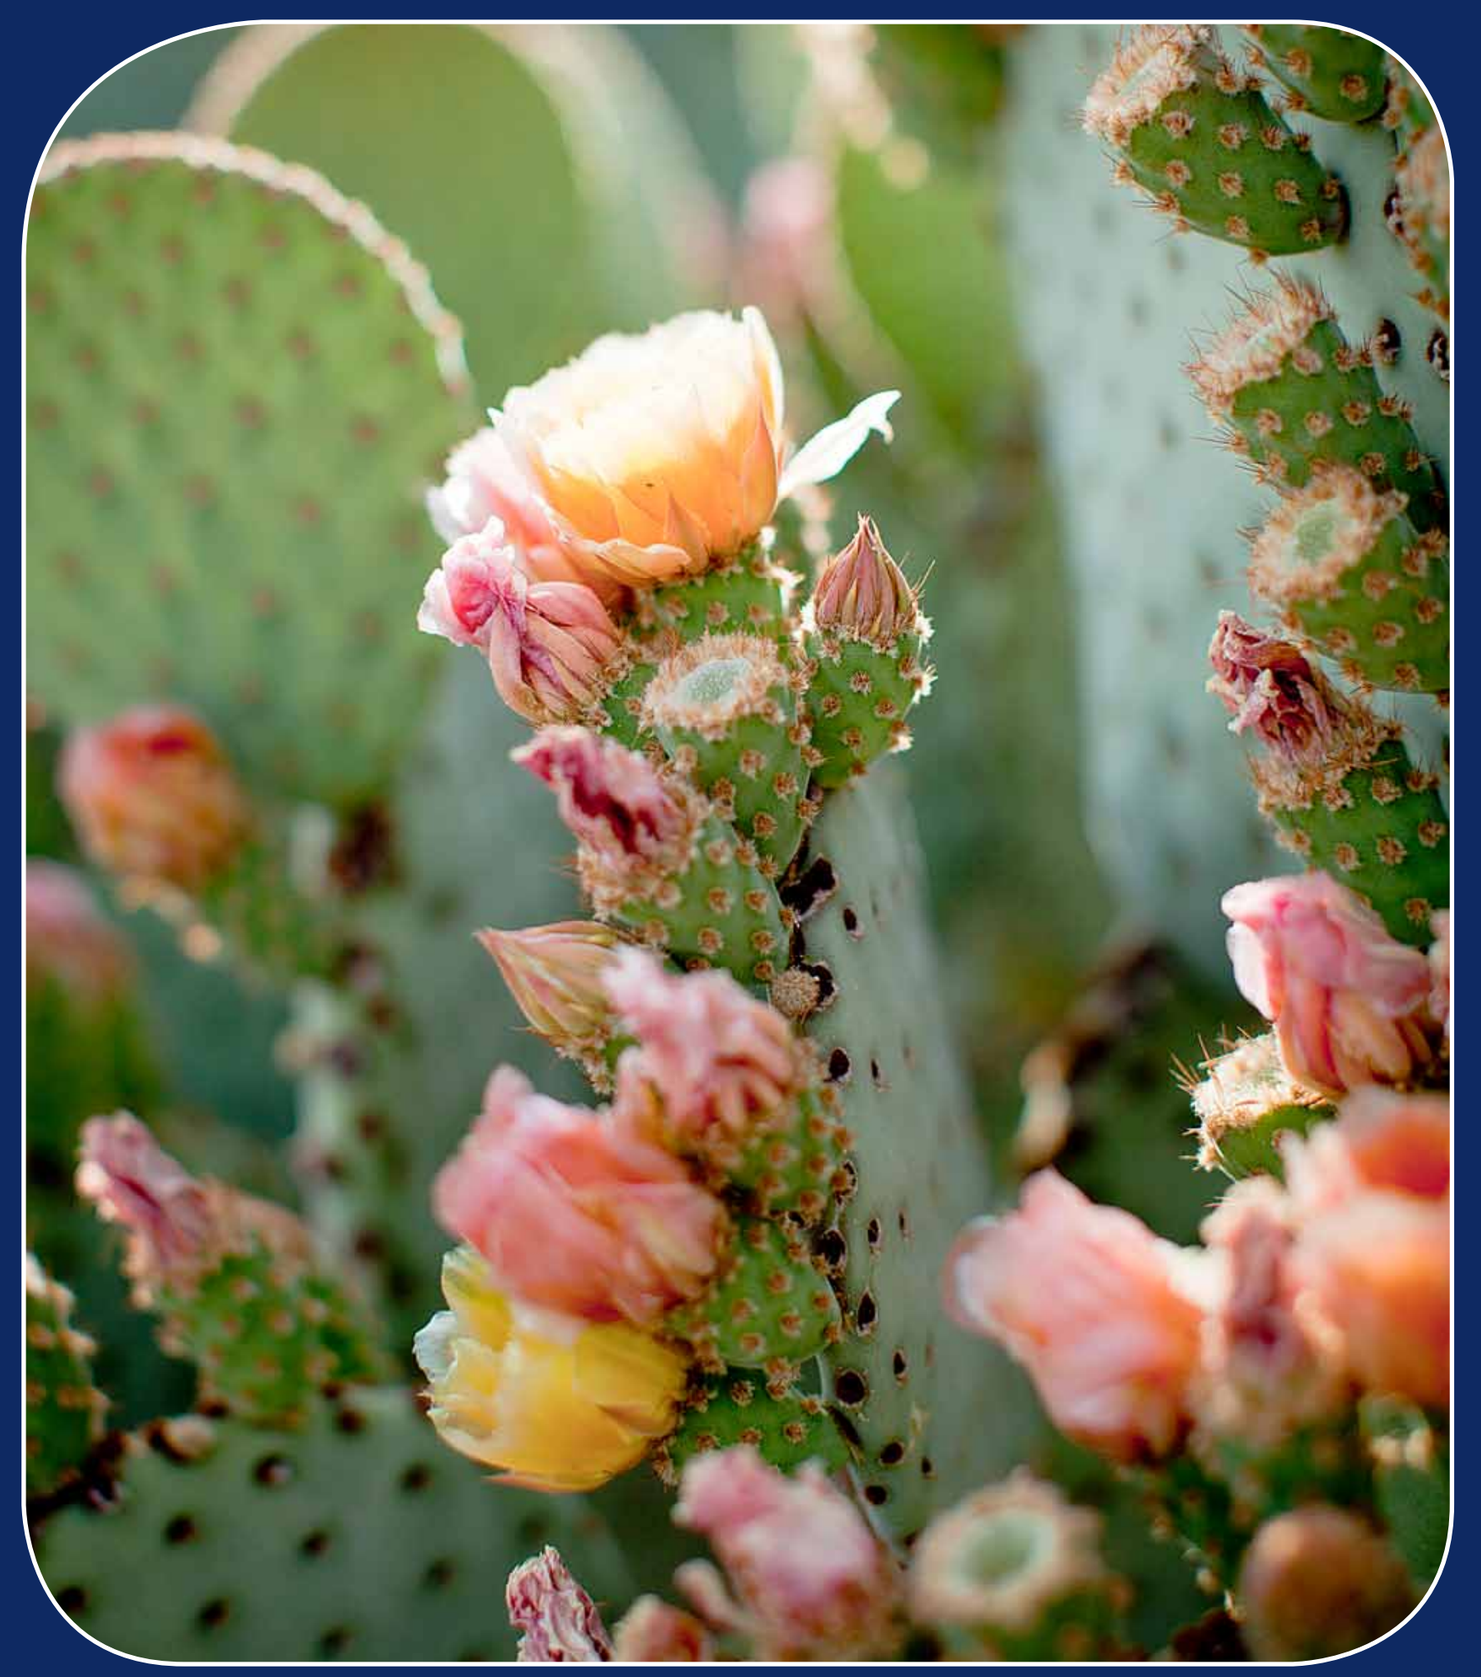

Acknowledgements
Katy Boeck, AAAAI

Funding Soruce
AAAAI

© 2013 Mayo Foundation for Medical Education and Research